Abstract number: 145

Phase 1 Efficacy and Pharmacodynamic Results of Exicorilant + Enzalutamide in Patients with Metastatic Castration-Resistant Prostate Cancer

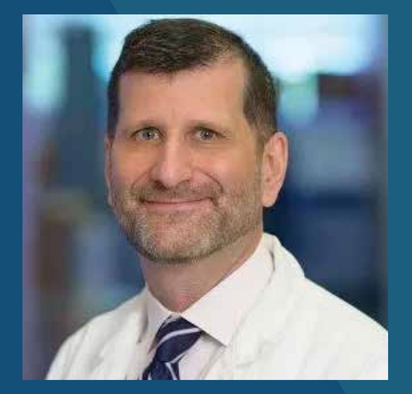

PRESENTER:
Michael J. Morris, MD
Contact: morrism@mskcc.org

Michael J. Morris[1], Mark Linch[2], Simon J. Crabb[3], Tomasz M. Beer[4], Elisabeth I. Heath[5], Michael S. Gordon[6], Johann de Bono[7], Hristina I. Pashova[8], Iulia Cristina Tudor[8], Andrew E. Greenstein[8], Grace Mann[8], Glenn Liu[9]

[1] Memorial Sloan Kettering Cancer Center, New York, NY; [2] University College London, London, UK; [3] Southampton Experimental Cancer Medicine Centre, University of Southampton, Southampton, Hampshire, UK; [4] OHSU Knight Cancer Institute, Portland, OR; [5] Karmanos Cancer Institute/Wayne State University, Detroit, MI; [6] HonorHealth Research & Innovation Institute, Scottsdale, AZ; [7] The Institute of Cancer Research and Royal Marsden Hospital, London, UK; [8] Concept Therapeutics Incorporated, Menlo Park, CA; [9] University of Wisconsin Carbone Cancer Center, Madison, WI

Summary & Conclusions

- There is an unmet need for novel, targeted treatments that can overcome tumor resistance pathways to existing therapies in mCRPC.
- This is the first study of the selective glucocorticoid receptor (GR) modulator (SGRM) exicorilant in combination with enzalutamide in patients with mCRPC.
- Exicorilant 240 mg QD + enzalutamide 160 mg QD was identified as a pharmacodynamically active regimen.[8]
- Modulation of GR target genes was confirmed at the exicorilant doses used in Segment 2.
- Despite pharmacodynamic activity, in the Segment 2 population of heavily pretreated patients receiving enzalutamide with rising PSA, significant drug activity, as reflected by PSA or radiographic responses, was not observed with the addition of exicorilant.
- Instances of PSADT improvements were predominantly observed in patients with higher baseline urinary free cortisol.
- Further study of exicorilant + enzalutamide in this advanced population is not currently planned.

The authors want to thank all those who participated in this study: The study patients and their families, the investigators, and the sponsor team.

Background

- Metastatic castration-resistant prostate cancer (mCRPC) remains an incurable disease with significant morbidity, despite the availability of multiple classes of therapies that delay disease progression and prolong life.[1]
- Androgen receptor (AR) signaling is a key driver of tumor growth in mCRPC.
 - AR-targeted therapies are the mainstay for patients with locally advanced or metastatic disease.
 - Enzalutamide (ENZA), an AR antagonist, is commonly used, but resistance typically develops within 8–12 months.[2-4]
- When AR is inhibited, prostate cancer cells can use the glucocorticoid receptor (GR) to activate AR-driven pathways, leading to AR antagonist resistance.[5,6]
- Combining the selective GR modulator (SGRM) exicorilant (EXI) with ENZA may block this escape pathway via dual antagonism of GR and AR.
 - EXI (CORT125281) is a competitive, reversible, full antagonist of the GR with high selectivity for GR relative to other hormone receptors.[7]
 - In the 22Rv1 CRPC xenograft model, EXI + ENZA reduced tumor growth,[8] supporting the hypothesis that dual antagonism of GR + AR may block this escape pathway.

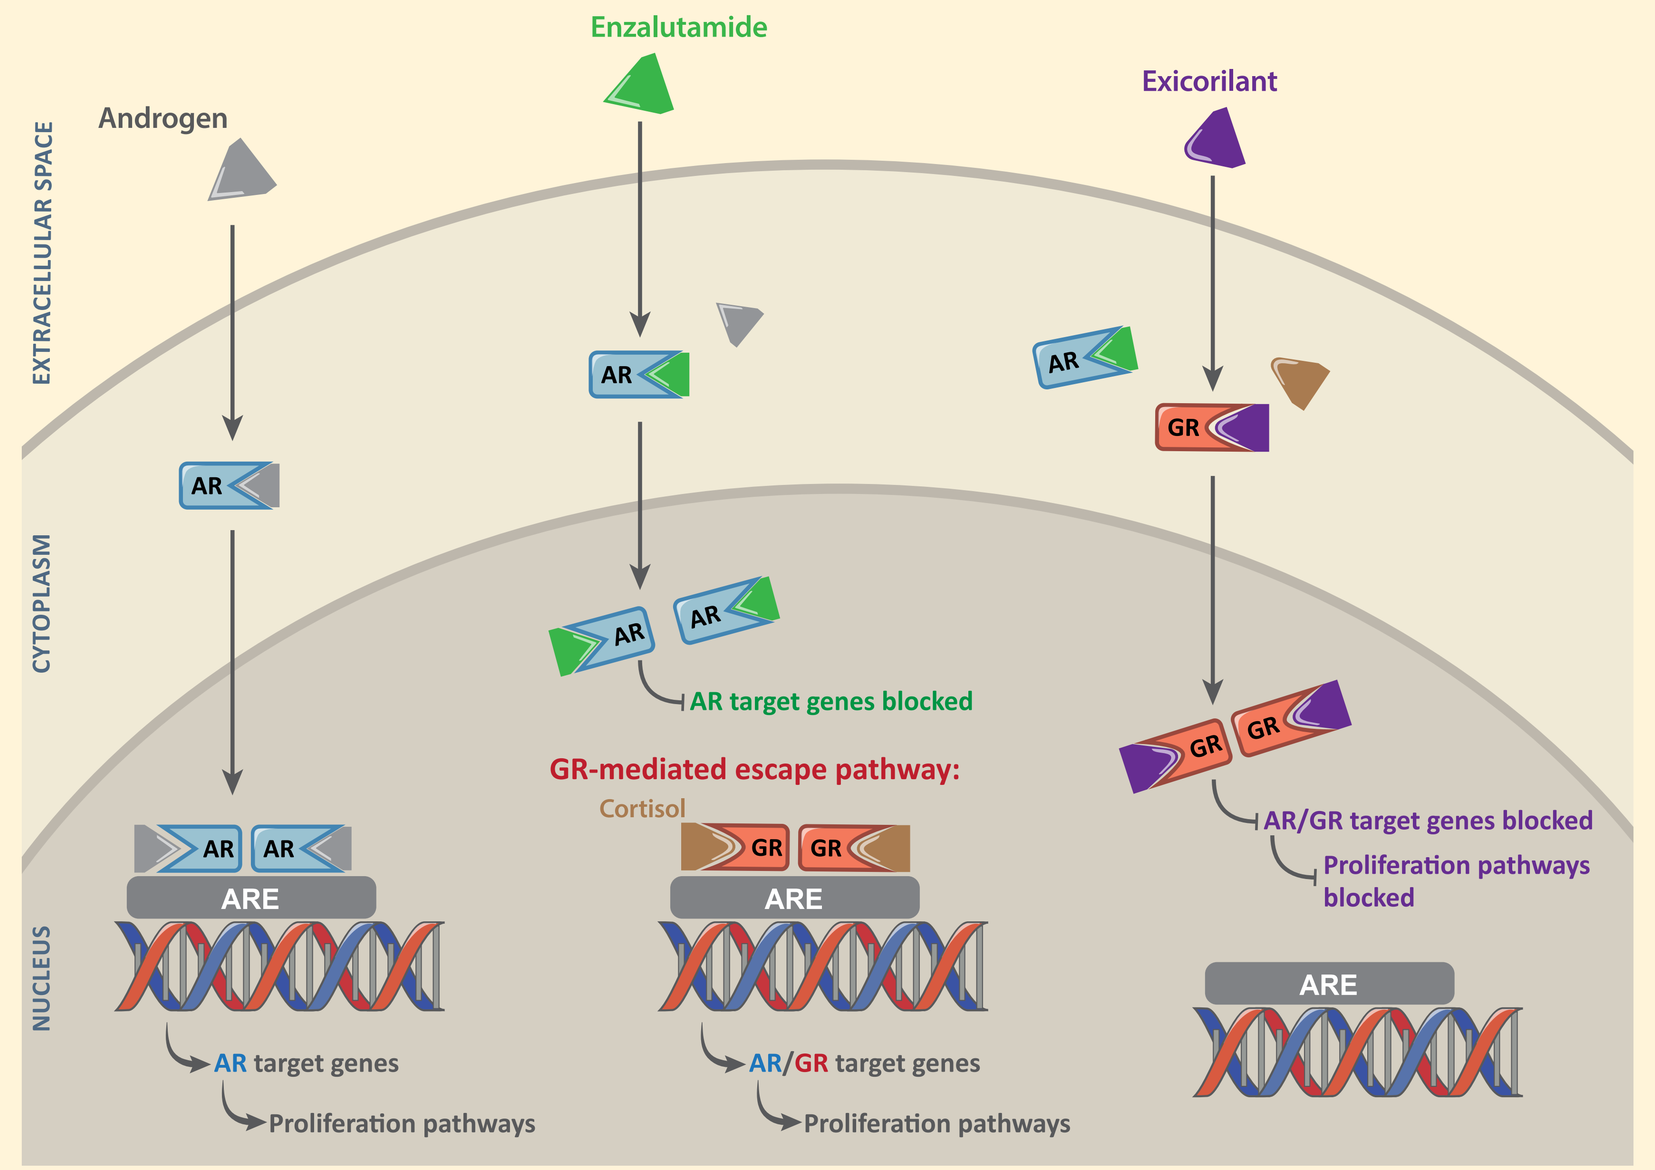

Androgen
Enzalutamide
Exicorilant
EXTRACELLULAR SPACE
CYTOPLASM
NUCLEUS
AR target genes blocked
GR-mediated escape pathway:
Cortisol
AR/GR target genes blocked
Proliferation pathways blocked
ARE
AR target genes
Proliferation pathways
AR/GR target genes

ARE, androgen response element.

- EXI + ENZA have been studied in a phase 1 study in patients with mCRPC (NCT03437941).
 - Safety and pharmacokinetic (PK) data were previously presented.[8]
 - Here, we report efficacy and biomarker results from this study.

1 Study Design

- **Patient population:** Men with histologically confirmed mCRPC who received ≤2 prior cytotoxic chemotherapy regimens
- **Segment 1:** EXI BID (140 or 180 mg, fasted) + ENZA 160 mg QD
 - Patients with progressive disease after most recent therapy (by imaging or PSA if PSA ≥1 ng/mL and rise confirmed by at least 2 measurements), irrespective of prior ENZA exposure.
 - Open-label '3+3' design, including 1 cohort with a 28-day ENZA monotherapy lead-in preceding cycle 1 of EXI + ENZA.
- **Segment 2:** EXI QD with food + ENZA at the currently tolerated, stable dose
 - Patients on a stable ENZA dose with rising PSA (25% increase over nadir and absolute value >1 ng/mL by at least 2 measurements)
 - Double-blind design: Patients received EXI 240 mg + ENZA and were randomized 3:1 to EXI titration (to 280 mg followed by 320 mg) or to remain on EXI 240 mg + placebo
- **Primary endpoint:** Determine a phase 2 regimen of EXI + ENZA*
- **Secondary & exploratory endpoints:** Safety*, PK*, efficacy of the combination, biomarkers
 - Efficacy assessments:
 - Imaging-based progression-free survival (PFS)
 - Changes in prostate-specific antigen (PSA) levels
 - PSA collected prior to the first EXI dose and PSA doubling times (PSADT) calculated before and during treatment in Segment 2
 - Biomarker analyses:
 - Baseline tumor GR expression (by CLIA-validated immunohistochemistry)
 - Modulation of GR target genes in whole blood (Paxgene; by a custom GR-targeted NanoString panel)
 - 24-h urinary free cortisol (UFC; by LC-MS/MS)
- **Data cutoff date:** July 7, 2022

* Primary endpoint, safety, and PK data presented at ESMO 2022.[8]

2 Baseline Characteristics

	Segment 1 (N=12)	Segment 2 (N=25)	Total (n=37)
Median age, years (range)	69.5 (53, 80)	71.0 (53, 82)	70.0 (53, 82)
ECOG performance status, n (%)			
0	8 (66.7%)	13 (52.0%)	21 (56.8%)
1	4 (33.3%)	12 (48.0%)	16 (43.2%)
Site of disease, n (%)			
Lung and/or liver	3 (25.0%)	5 (20.0%)	8 (21.6%)
Bone only	3 (25.0%)	12 (48.0%)	15 (40.5%)
Median PSA level, ng/mL (range)	11.2 (0.1, 350)	16.3 (3.2, 2300)	15.1 (0.1, 2300)
Median time since pathological diagnosis, years	8.8	4.4	5.2
Median Gleason score at diagnosis, n (range)	8 (7, 10)	8 (6, 10)	8 (6, 10)
8-10, n (%)	10 (83.3%)	16 (64.0%)	26 (70.3%)
Prior AR pathway inhibitor, n (%)	5 (41.7%)	25 (100%)	30 (81.1%)
Abiraterone	5 (41.7%)	6 (24.0%)	11 (29.7%)
Apalutamide	1 (8.3%)	1 (4.0%)	2 (5.4%)
Enzalutamide	1 (8.3%)	25 (100%)	26 (70.3%)
Nilutamide	1 (8.3%)	0	1 (2.7%)
Prior taxane, n (%)	6 (50.0%)	13 (52.0%)	19 (51.4%)
Cabazitaxel	3 (25.0%)	1 (4.0%)	4 (10.8%)
Docetaxel	6 (50.0%)	12 (48.0%)	18 (48.6%)

Two enrolled patients did not receive EXI and are not included in this table. One patient enrolled in Segment 2 did not have soft-tissue or bone disease at baseline.

3 Biomarker Results

Exicorilant Doses in Segment 2 Achieved GR Modulation

- 8 GR target genes were validated as specific biomarkers of the SGRM relacorilant in a separate randomized phase 2 ovarian cancer study (NCT03776812).
- Modulation of these genes at EXI doses of 240–320 mg QD for 2 weeks with food (Segment 2) demonstrated a specific PD effect.
 - CDKN1C, TNFRSF17, BRIP1, and PDK1 were suppressed; LILRB4, FPR3, CLEC10A, and CCR2 were increased after 2 weeks at each dose.
- Fasting BID dosing of EXI in Segment 1 was significantly less active.
- Enzalutamide alone did not alter these genes.

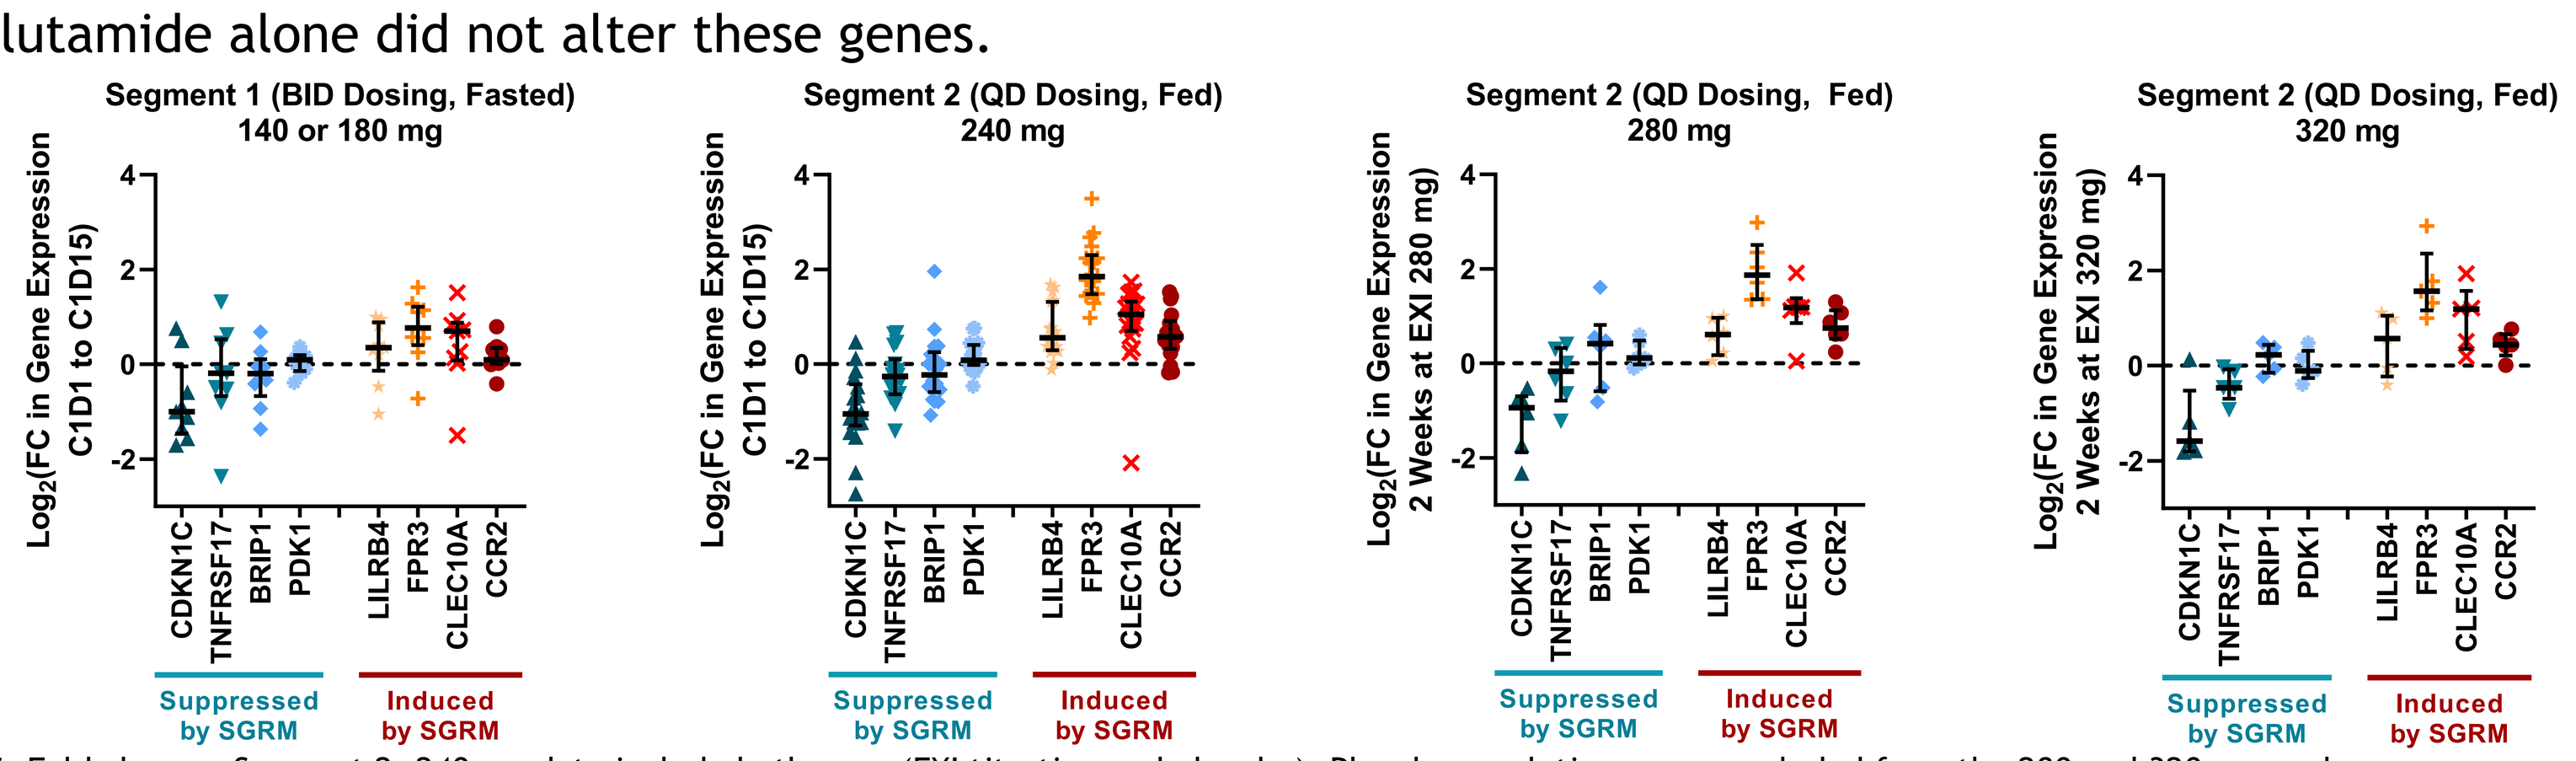

Segment 1 (BID Dosing, Fasted) 140 or 180 mg
Segment 2 (QD Dosing, Fed) 240 mg
Segment 2 (QD Dosing, Fed) 280 mg
Segment 2 (QD Dosing, Fed) 320 mg
Log_2(FC in Gene Expression C1D1 to C1D15)
Log_2(FC in Gene Expression 2 Weeks at EXI 280 mg)
Log_2(FC in Gene Expression 2 Weeks at EXI 320 mg)
CDKN1C
TNFRSF17
BRIP1
PDK1
LILRB4
FPR3
CLEC10A
CCR2
Suppressed by SGRM
Induced by SGRM

FC, Fold change. Segment 2: 240 mg data include both arms (EXI titration and placebo). Placebo escalations were excluded from the 280 and 320 mg analyses.

Exicorilant Did Not Affect Cortisol Levels

- While treatment with the non-selective GR antagonist mifepristone typically results in 3-fold elevations in 24-h UFC[9], selective GR modulation with EXI did not alter cortisol levels.
- Similarly, serum cortisol and adrenocorticotropic hormone were not significantly altered by EXI.[8]
- Thus, PD, efficacy, and safety analyses can assess GR modulation by EXI without complication from increased agonist (cortisol) levels.

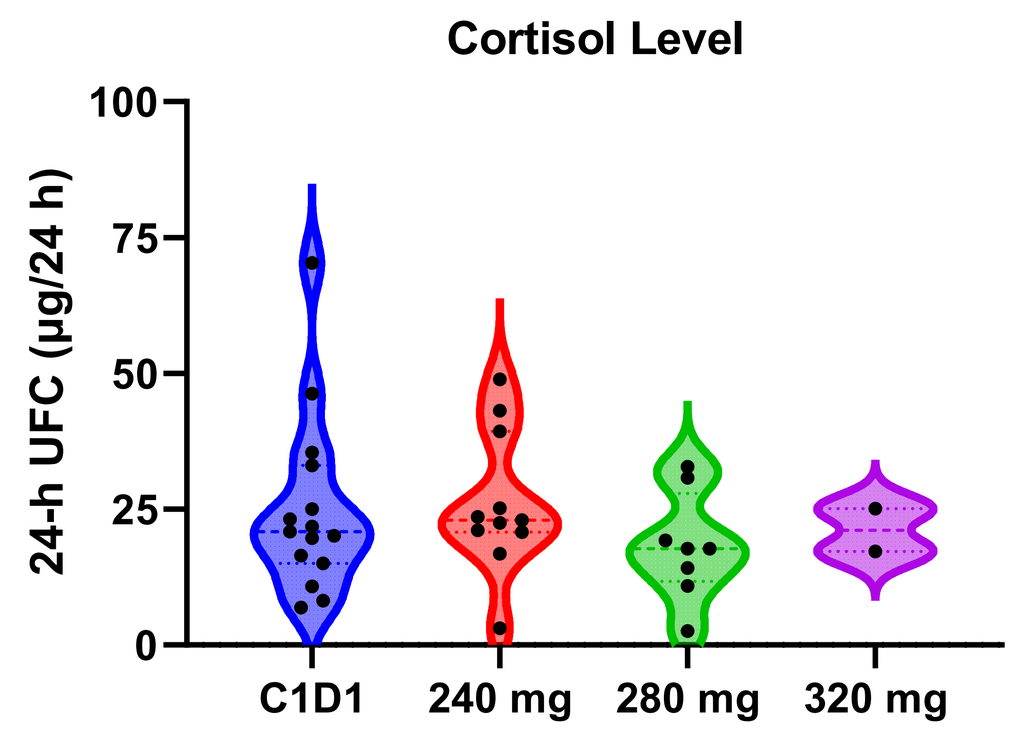

Cortisol Level
24-h UFC (μg/24 h)
C1D1
240 mg
280 mg
320 mg

Figure shows only Segment 2 data in subjects who escalated on EXI. 24-h UFC reference range: 10-100 μg/day. C1D1, cycle 1 day 1.

Higher UFC Was Associated with a SGRM PD Marker and PSADT Increases After Treatment with EXI + ENZA (Segment 2)

- Baseline expression of CLEC10A, a marker of GR activity, in blood was associated with baseline 24-h UFC.
 - Higher cortisol levels correspond to lower CLEC10A (left Figure); SGRM treatment increases CLEC10A (see above).
- PSADT increases after treatment with EXI + ENZA were predominantly observed in patients with higher baseline 24-h UFC (*P*<0.05; right Figure).
 - Low baseline UFC was associated with PSADT decreases on study treatment.
 - In most patients, baseline UFC was within the normal range (3.5–45 μg/24 h).
 - Limitations include the small sample size and limited number of responses in this heavily pretreated population.

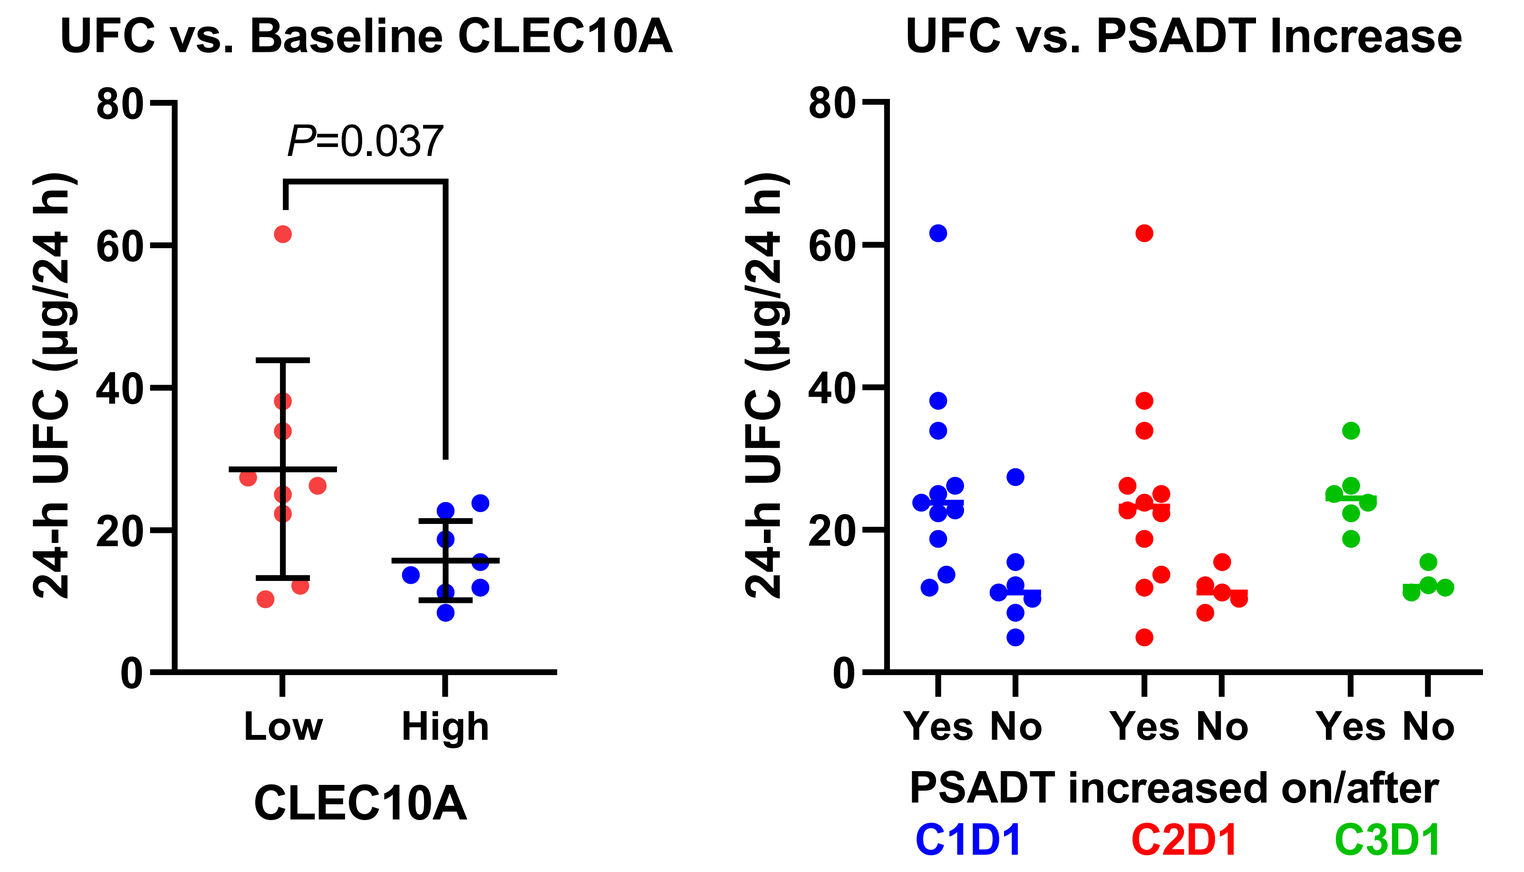

UFC vs. Baseline CLEC10A
P=0.037
24-h UFC (μg/24 h)
Low
High
CLEC10A
UFC vs. PSADT Increase
Yes No
PSADT increased on/after
C1D1
C2D1
C3D1

GR Expression Was Detectable in all Assessed Baseline Tumor Samples

- High levels of nuclear GR immunoreactivity[10] were observed in nearly all evaluable tumor specimens (n=32), confirming high GR expression in mCRPC patients resistant to AR antagonist (ie, with rising PSA on ENZA).

Images on the right show GR nuclear staining in 2 patient samples.

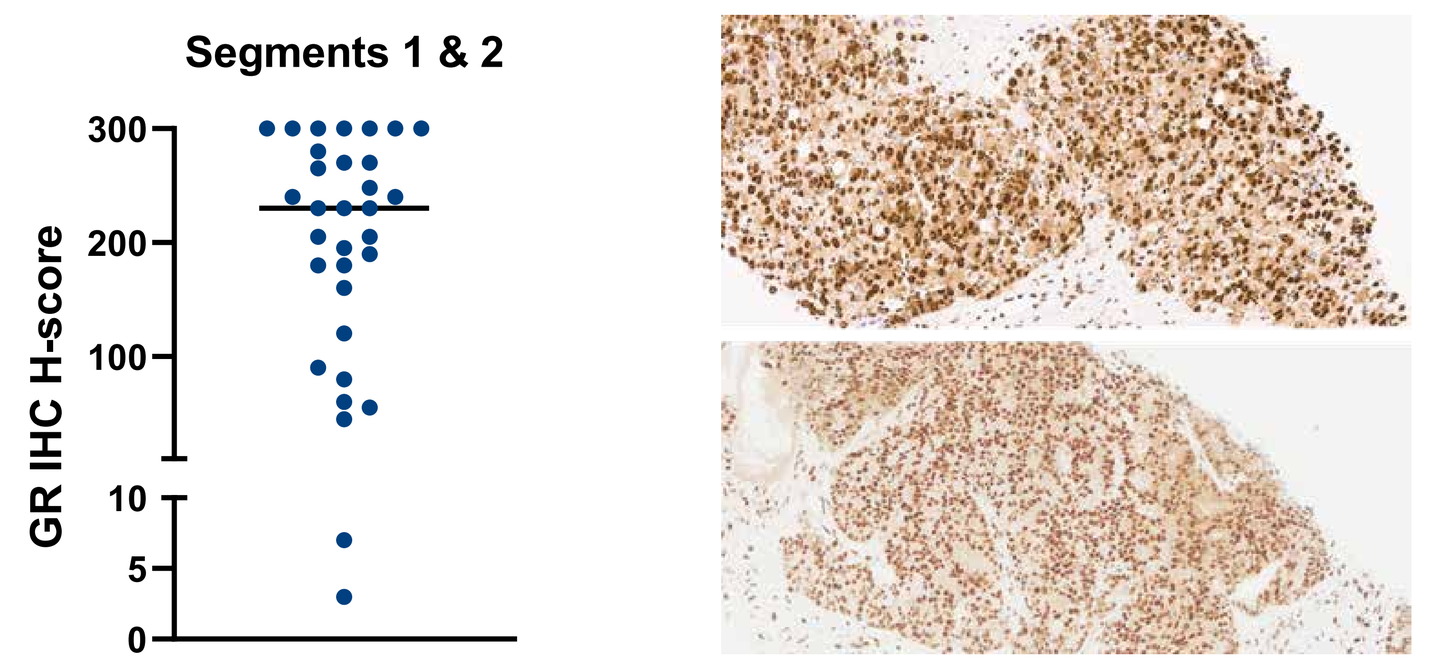

Segments 1 & 2
GR IHC H-score

4 Efficacy Results (Segment 2)

Best Overall Response and Progression-free Survival

- As QD dosing of EXI under fed conditions achieved GR modulation, efficacy results focus on Segment 2.
- There were no imaging-based tumor responses per PCWG3/mRECIST v1.1.
- 18 patients had a best overall response of stable disease per PCWG3/mRECIST v1.1.
- Median duration of exposure to EXI was 9.7 weeks (range: 2-61).

	Segment 2 (N=25)
Best overall response*, n (%)	
Complete response (CR)	0
Partial response (PR)	0
Stable disease (SD)	18 (72.0%)
Progressive disease (PD)	4 (16.0%)
Not evaluable	1 (4.0%)
Not assessed	2 (8.0%)
Median imaging-based PFS†, months (95% CI)	5.5 (3.7, –)
Median PFS follow-up time, months (95% CI)	4.6 (1.7, 8.3)

* Tumor response assessed by PCWG3, incorporating modified RECIST v1.1. † Imaging-based PFS assessed by mRECIST v1.1, progression on bone lesions per PCWG3, or death.
PCWG3, Prostate Cancer Clinical Trials Working Group 3; RECIST, Response Evaluation Criteria in Solid Tumors.

Modest PSA Reductions Were Observed

- PSA reductions from baseline occurred in 4/25 patients (16%).
 - 1 PSA reduction >50% from baseline, confirmed 4 weeks later, was observed at EXI 320 mg + ENZA 160 mg (max. PSA reduction: 71.1%).
 - 1 PSA reduction >25% from baseline, confirmed 4 weeks later, was observed at EXI 240 mg + ENZA 160 mg (max. PSA reduction: 36.7%).

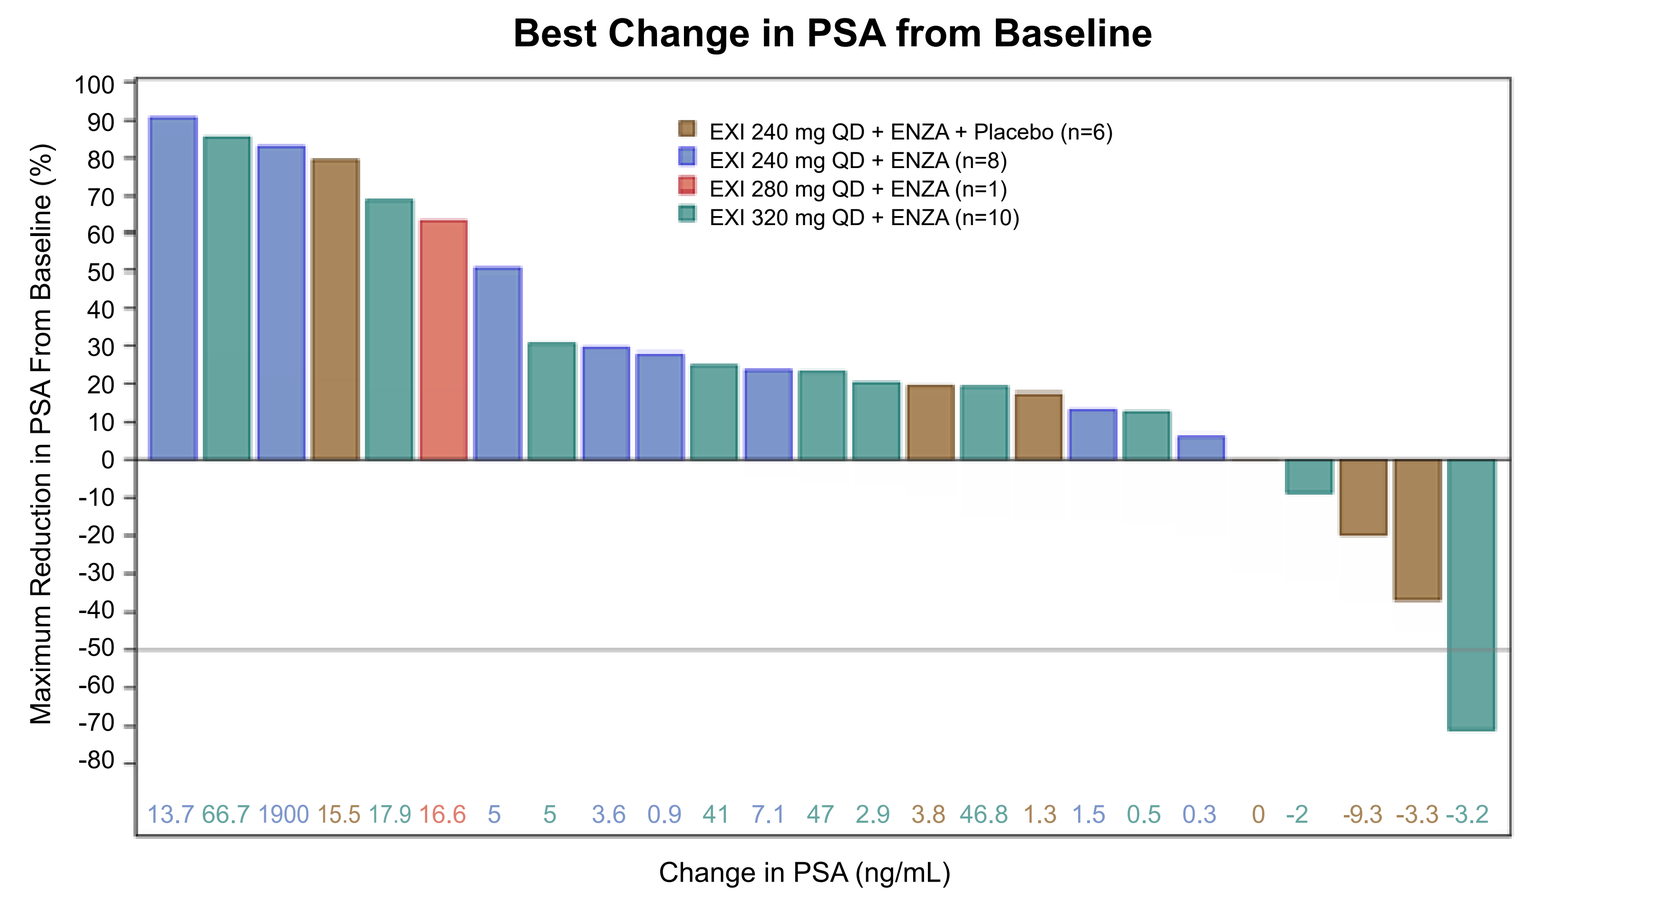

Best Change in PSA from Baseline
EXI 240 mg QD + ENZA + Placebo (n=6)
EXI 240 mg QD + ENZA (n=8)
EXI 280 mg QD + ENZA (n=1)
EXI 320 mg QD + ENZA (n=10)
Maximum Reduction in PSA From Baseline (%)
13.7 66.7 1900 15.5 17.9 16.6 5 5 3.6 0.9 41 7.1 47 2.9 3.8 46.8 1.3 1.5 0.5 0.3 0 -2 -9.3 -3.3 -3.2
Change in PSA (ng/mL)

- PSA doubling times (PSADT) increased after study treatment in 13/25 patients (52%).
 - Among those who received more than 2 cycles of treatment, 61.5% had a PSADT increase.

- PSA reductions during treatment, despite rising PSA at study entry, were observed in 14/25 patients (56%, shown in purple in the figure below).
 - In 11/14 patients, PSA declines occurred by the second on-treatment PSA assessment.

* Note: PSA declines for 3 patients occurred after day 84 and aren't visible in the figure on the right.

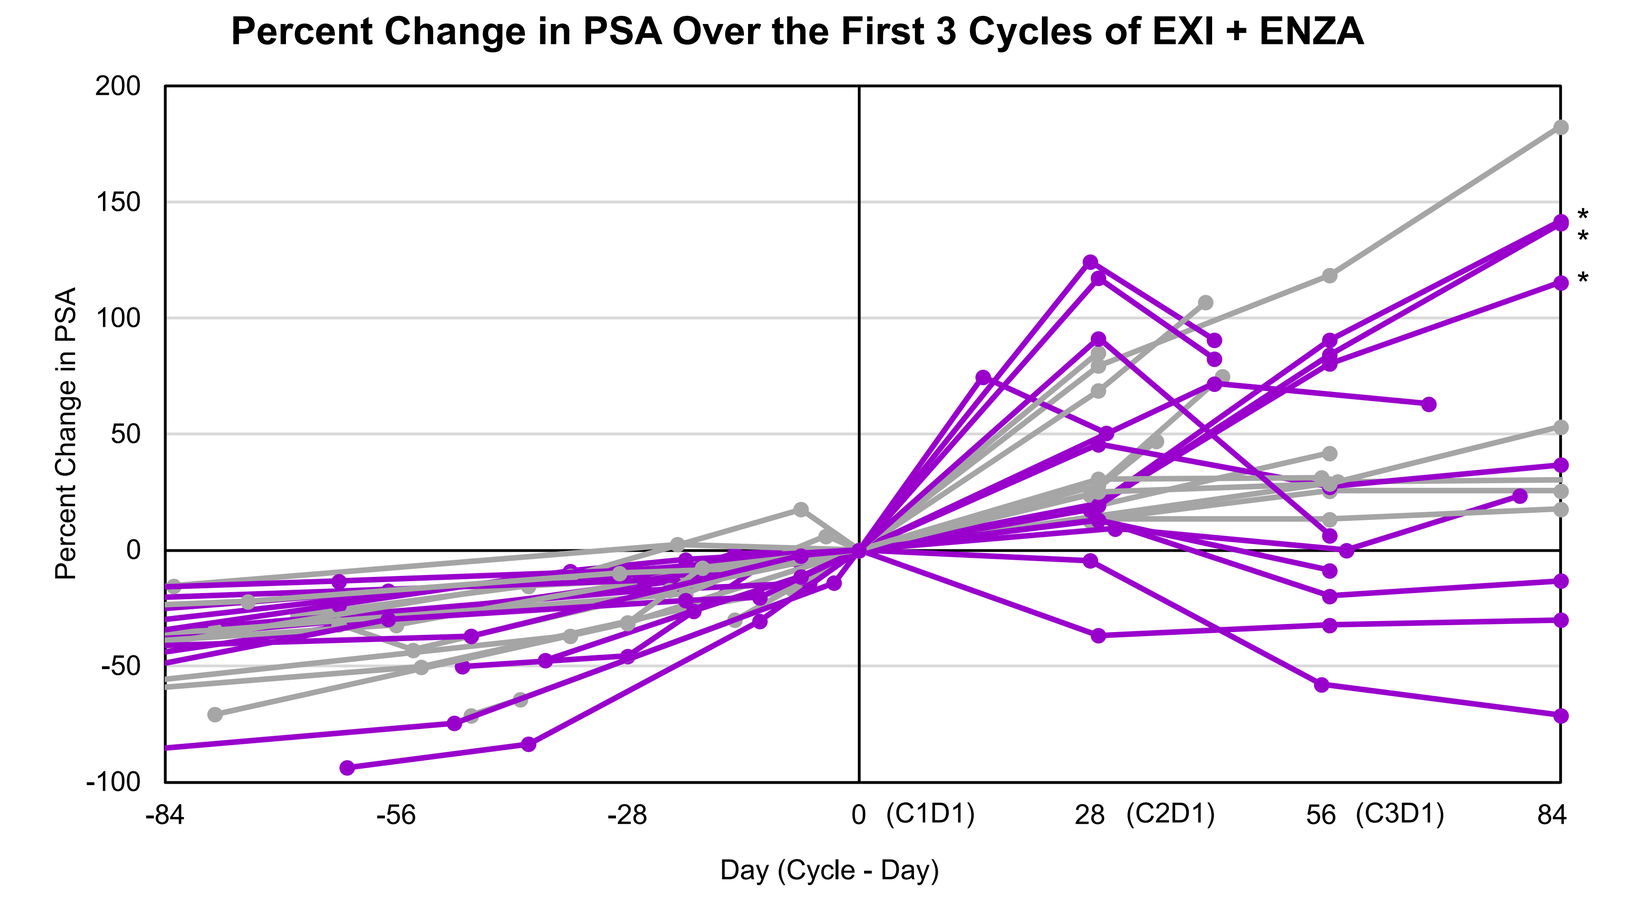

Percent Change in PSA Over the First 3 Cycles of EXI + ENZA
Percent Change in PSA
0 (C1D1)
28 (C2D1)
56 (C3D1)
Day (Cycle - Day)

Segment 2 (N=25)		
	Pre-treatment median PSADT, months	3.6
	PSADT increased, n (%)	
	After C1D1 (n=25)	13 (52.0%)
	After C2D1 (n=23)	16 (69.6%)
	After C3D1 (n=13)	8 (61.5%)

PSADT was calculated prior to the first EXI dose (on ENZA alone), after C1D1, C2D1, and C3D1.

References

1. Henriquez et al. *Biomedicines*. 2021;9(9):1247.
2. Tran et al. *Science*. 2009;324 (5928):787.
3. Scher et al. *NEJM*. 2012;367: 1187.
4. Beer et al. *NEJM*. 2014;371:424.
5. Arora et al. *Cell*. 2013;155(6): 1309.
6. Isikbay et al. *Horm Cancer*. 2014;5(2):72.
7. Hunt et al. *J Med Chem*. 2017; 60(8):3405.
8. Linch et al. *Ann Oncol*. 2022;33(suppl_7):S616.
9. Gubbi et al. *J Clin Endocrinol Metab*. 2021;106(5):1501.
10. Block et al. *Cancer Manag Res*. 2017;9:65.

Acknowledgements

This study is sponsored by Corcept Therapeutics. Medical writing assistance was provided by Tina Schlafly, PhD, CMPP, of Corcept Therapeutics.

Disclosures

Please see ASCO website for authors' disclosures.