A STUDY OF RELACORILANT IN COMBINATION WITH NAB-PACLITAXEL IN PATIENTS WITH METASTATIC PANCREATIC DUCTAL ADENOCARCINOMA (mPDAC)

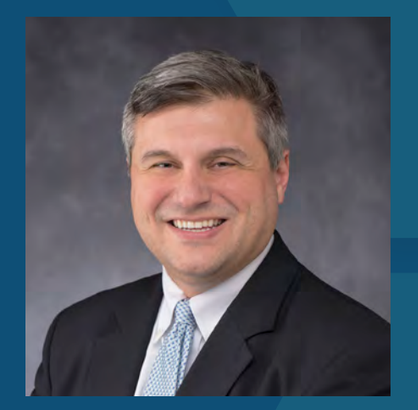

PRESENTER:
Erkut H. Borazanci, MD
Contact: eborazanci@honorhealth.com

Erkut H. Borazanci, MD[1]; Nathan Bahary, MD, PhD[2]; Vincent Chung, MD[3]; Roland T. Skeel, MD[4]; E. Gabriela Chiorean, MD[5]; Andrew Greenstein, PhD[6]; Hristina Pashova, PhD[6]; Iulia Cristina Tudor, PhD[6]; Grace Mann, PhD[6]; Paul E. Oberstein, MD[7]

[1] HonorHealth Research Institute, TGen, Scottsdale, AZ, USA; [2] University of Pittsburgh, Pittsburgh, PA, USA (current affiliation: Allegheny Health Network Cancer Institute, Pittsburgh, PA, USA); [3] City of Hope Comprehensive Cancer Center, Duarte, CA, USA; [4] The University of Toledo Medical Center, Toledo, OH, USA; [5] University of Washington, Fred Hutchinson Cancer Research Center, Seattle, WA, USA; [6] Corcept Therapeutics, Menlo Park, CA, USA; [7] New York University Langone Health, New York, NY, USA

Conclusions

- **No new safety signals were observed with relacorilant + nab-paclitaxel.**
- **Whole blood RNA analysis confirmed that the combination of relacorilant + nab-paclitaxel suppresses glucocorticoid receptor (GR) target genes, similar to the findings in other tumor types.**
- **Modest antitumor activity of relacorilant + nab-paclitaxel was observed in this study of heavily pretreated patients with mPDAC.**
- **While relacorilant + nab-paclitaxel showed antitumor activity, the apparent level of benefit did not justify further study as a treatment for end-stage pancreatic cancer.**
- **Relacorilant + nab-paclitaxel continues to be evaluated for the treatment of other tumor types.**

The authors want to thank all those who participated in this study: the study patients and their families, the investigators, and the sponsor team.

ASCO 2022
Presented at the American Society of Clinical Oncology Annual Meeting; June 3-7, 2022; Chicago, IL

Background

- Pancreatic cancer remains the third leading cause of cancer-related death in the United States.[1]
 - The average overall survival (OS) after diagnosis of advanced, metastatic disease is only 1 year.[2,3]
 - (Modified) FOLFIRINOX or nab-paclitaxel and gemcitabine lead to only modest improvements to OS.[2,3]
 - Third-line treatment options for mPDAC are limited, with response rates near 0%.[2]
- Chemotherapy resistance is a major concern in the treatment of pancreatic cancer.[4]
 - Most patients ultimately succumb to the disease due to the emergence of chemotherapy resistance.[4]
 - One mechanism of resistance is driven by cortisol, which can inhibit drug-induced apoptosis by binding to the GR, activating tumor survival genes such as SGK1 and DUSP1.[5,6]
- Preclinical and clinical data indicate that modulation of GR signaling can reverse the anti-apoptotic effects of cortisol, thereby enhancing the efficacy of cytotoxic agents.[7]
 - **Relacorilant** is a reversible, selective GR modulator (SGRM) that has shown promise in resensitizing tumors to taxanes in preclinical models in various solid tumors.[7]
 - In a phase 2 study in patients with ovarian cancer, relacorilant + nab-paclitaxel showed improved progression-free survival (PFS) without increased side effects compared to nab-paclitaxel alone.
- Here, we report the results of RELIANT, a trial evaluating the efficacy and safety of relacorilant with nab-paclitaxel in patients with mPDAC.

1 RELIANT Study Design

- **RELIANT** (NCT04329949) was a single-arm, open-label, multicenter study of relacorilant (100 mg once daily titrating up to 150 mg in 25 mg increments) + nab-paclitaxel (80 mg/m² on Days 1, 8, and 15 of each 28-day cycle) in patients with histologically confirmed mPDAC.
 - Relacorilant inhibits the CYP3A4-mediated metabolism of nab-paclitaxel, resulting in increased nab-paclitaxel exposure when co-administered.[8] Nab-paclitaxel 80 mg/m² combined with relacorilant approximates the exposure achieved with 100 to 125 mg/m² nab-paclitaxel alone.[9]
 - Prophylactic G-CSF administration was required to reduce neutropenia risk.
- Patients with mPDAC who had received ≥2 prior lines of therapy for PDAC in any setting, including prior gemcitabine-based therapy and fluoropyrimidine-based therapy, were enrolled.
- Planned enrollment was approximately 80 patients.
- The study included a planned interim analysis after approximately 40 patients had completed 12 weeks of treatment or discontinued due to disease progression or toxicity.
 - The interim analysis included 43 patients (with data cutoff date of May 25, 2021).
 - Objective response rate (ORR) by investigator was used to determine whether to continue or stop enrollment after the interim analysis.
- The primary endpoint of the study was ORR as assessed by blinded independent central review.
 - Key secondary endpoints include ORR by investigator, duration of response (DOR), PFS, OS, and assessment of cancer antigen 19-9 (CA19-9) in patients with elevated CA19-9 at baseline.

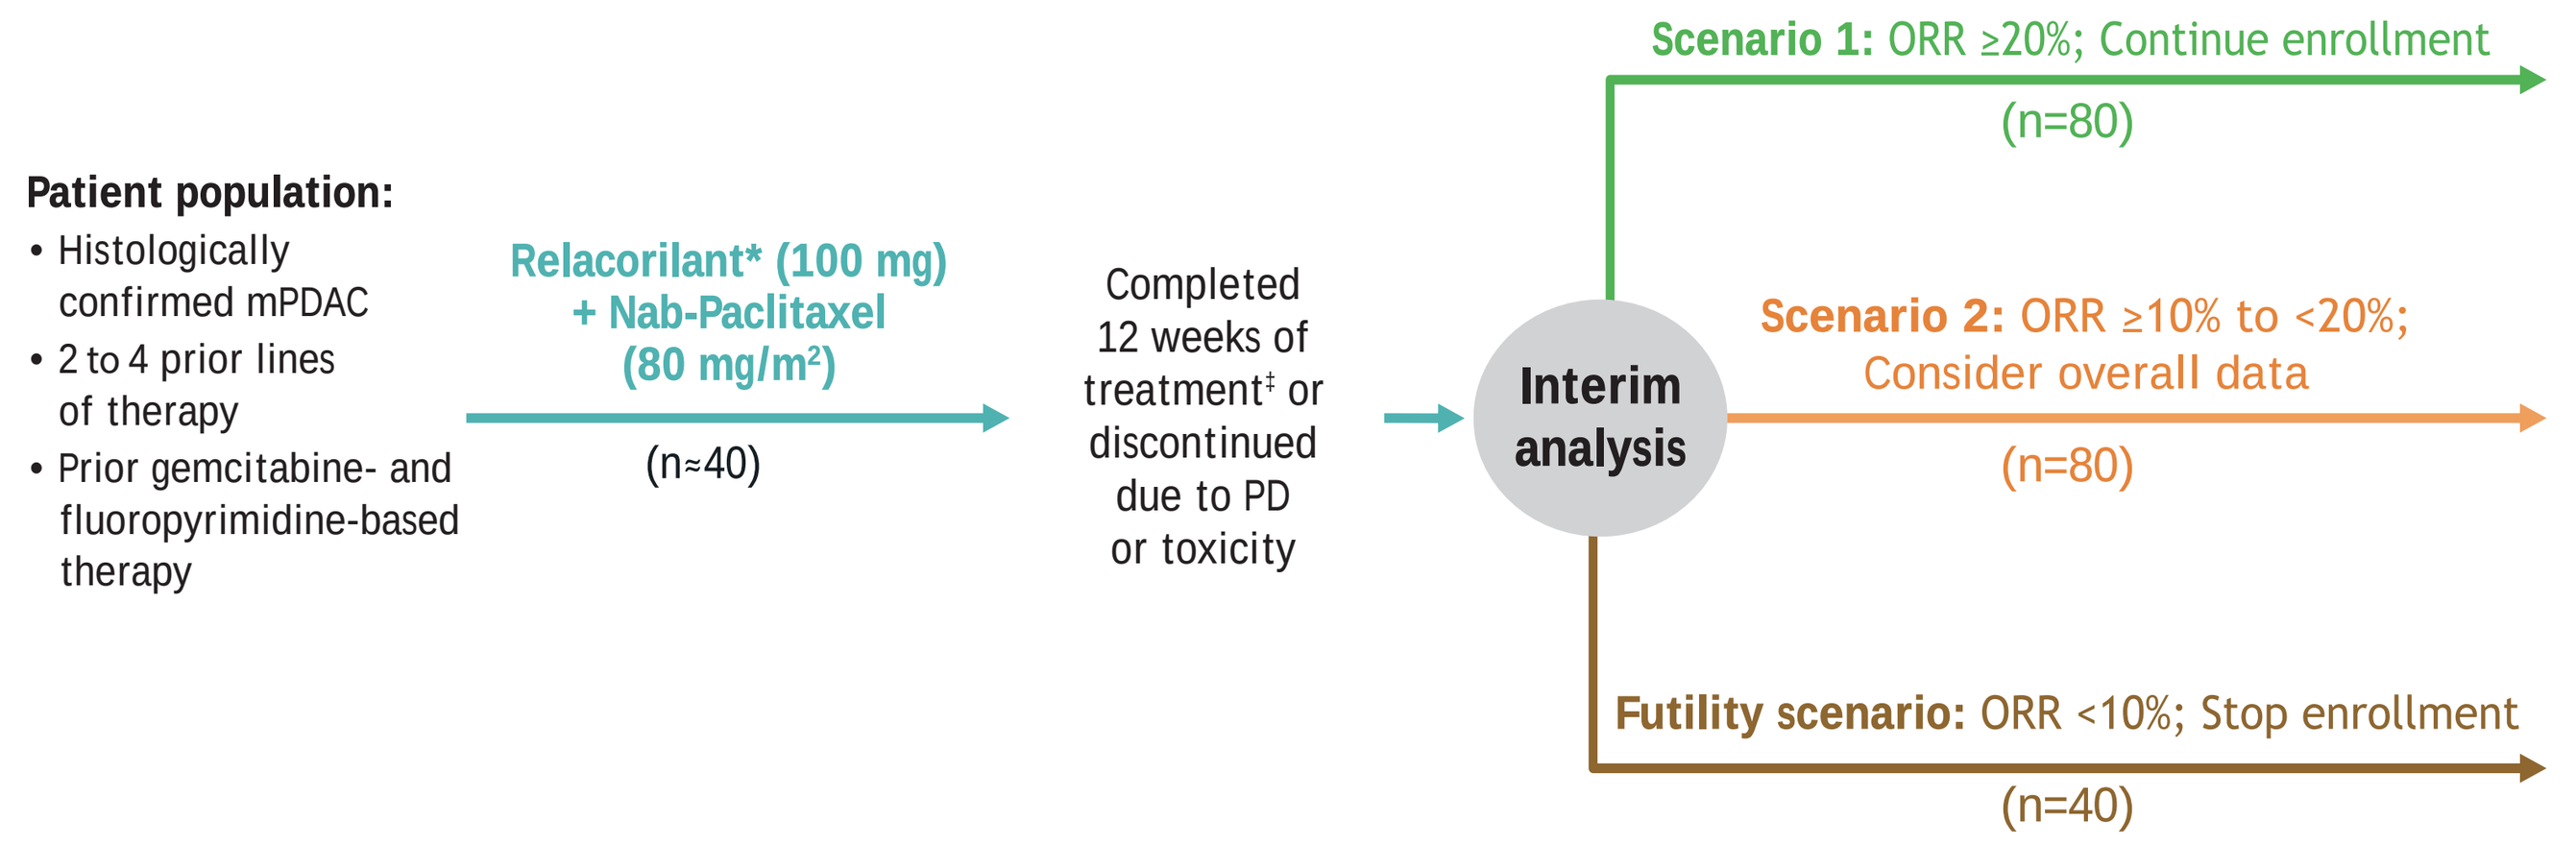

Patient population:
• Histologically confirmed mPDAC
• 2 to 4 prior lines of therapy
• Prior gemcitabine- and fluoropyrimidine-based therapy
Relacorilant* (100 mg) + Nab-Paclitaxel (80 mg/m²)
(n≈40)
Completed 12 weeks of treatment‡ or discontinued due to PD or toxicity
Interim analysis
Scenario 1: ORR ≥20%; Continue enrollment
(n=80)
Scenario 2: ORR ≥10% to <20%; Consider overall data
(n=80)
Futility scenario: ORR <10%; Stop enrollment
(n=40)

*Starting 100 mg once daily with titration up to 150 mg once daily in 25 mg increments.
‡Including second radiographic assessment and ≥1 post-baseline tumor assessment other than non-evaluable.
G-CSF, granulocyte colony-stimulating factor; PD, progressive disease.

2 Baseline Demographics and Disposition

- A total of 43 patients were enrolled.
- As of April 15, 2022, all patients have discontinued the study.
- The most common reasons for discontinuation from relacorilant were disease progression (n=17, 39.5%), adverse event (AE; n=10, 23.3%), and patient decision (n=8, 18.6%).

	Relacorilant + nab-paclitaxel (N=43)
Age, median (range), y	64 (43, 78)
<65, n (%)	22 (51.2)
≥65, n (%)	21 (48.8)
Sex, male, n (%)	24 (55.8)
Ethnicity, n (%)	
Hispanic or Latino	3 (7.0)
Not Hispanic or Latino	37 (86.0)
Not reported	3 (7.0)
Number of prior therapy lines	
2	16 (37.2)
3	17 (39.5)
≥4	10 (23.3)
Number of prior therapies in metastatic setting, median (range)	3 (2, 4)
In any setting	3 (2, 5)
Prior therapy in any setting, n (%)	
Prior gemcitabine and prior fluoropyrimidine	43 (100.0)
No prior nab-paclitaxel	3 (7.0)
Of patients with no prior nab-paclitaxel, other prior taxane	1 (33.3)

3 Antitumor Activity Observed in a Heavily Pretreated Patient Population

- In the efficacy-evaluable population, 17/31 (54.8%) patients had a best overall resonse of CR or PR.
 - In these end-stage pancreatic cancer patients, 12/43 (28%) did not reach the first post-baseline radioagraphic assessment
- The ORR by investigator did not meet the predefined threshold of ≥10%, and enrollment was stopped after the interim analysis (May 25, 2021).

Efficacy endpoints for RELIANT in the evaluable population

Investigator assessment per RECIST v1.1	ITT population (N=43)	Evaluable population (N=31)
ORR (confirmed), n (%)	0	0
Best overall response (confirmed or unconfirmed), n (%)		
CR	0	0
PR	2 (4.7)	2 (6.5)
SD	15 (34.9)	15 (48.4)
PD	14 (32.6)	14 (45.1)
Non-evaluable	12 (27.9)	0
Disease control rate,* n (%)		
At 6 weeks	16 (37.2)	16 (51.6)
At 12 weeks	10 (23.3)	10 (32.3)
At ≥18 weeks	5 (11.6)	5 (16.1)
PFS, median (95% CI), months	1.6 (1.4, 3.1)	N/A
OS, median (95% CI), months	3.9 (2.8, 4.9)	N/A

The ITT population included all enrolled patients who were treated with ≥1 dose of combination treatment. The evaluable population included all patients in the ITT population who had ≥1 post-baseline radiographic tumor assessment available (with results other than non-evaluable). *Defined as the percentage of patients with confirmed or unconfirmed CR or PR, or SD at 6, 12, or ≥18 weeks. CI, confidence interval; CR, complete response; ITT, intent-to-treat; N/A, not available; PR, partial response; RECIST, Response Evaluation Criteria in Solid Tumors; SD, stable disease.

- Relacorilant + nab-paclitaxel demonstrated antitumor activity, with 15/32 (46.9%) patients with baseline and post-baseline assessments of target lesions showing decreases in target lesion size**.**
 - Patients remained on treatment for 1 to 17 cycles.
- In 17/39 (43.5%) patients with elevated CA19-9 at baseline, CA19-9 decreased; in 5/39 (12.8%) the decrease was >50%.

Waterfall plot of best percent change from baseline in target lesions in patients with a post-baseline target lesion measurement

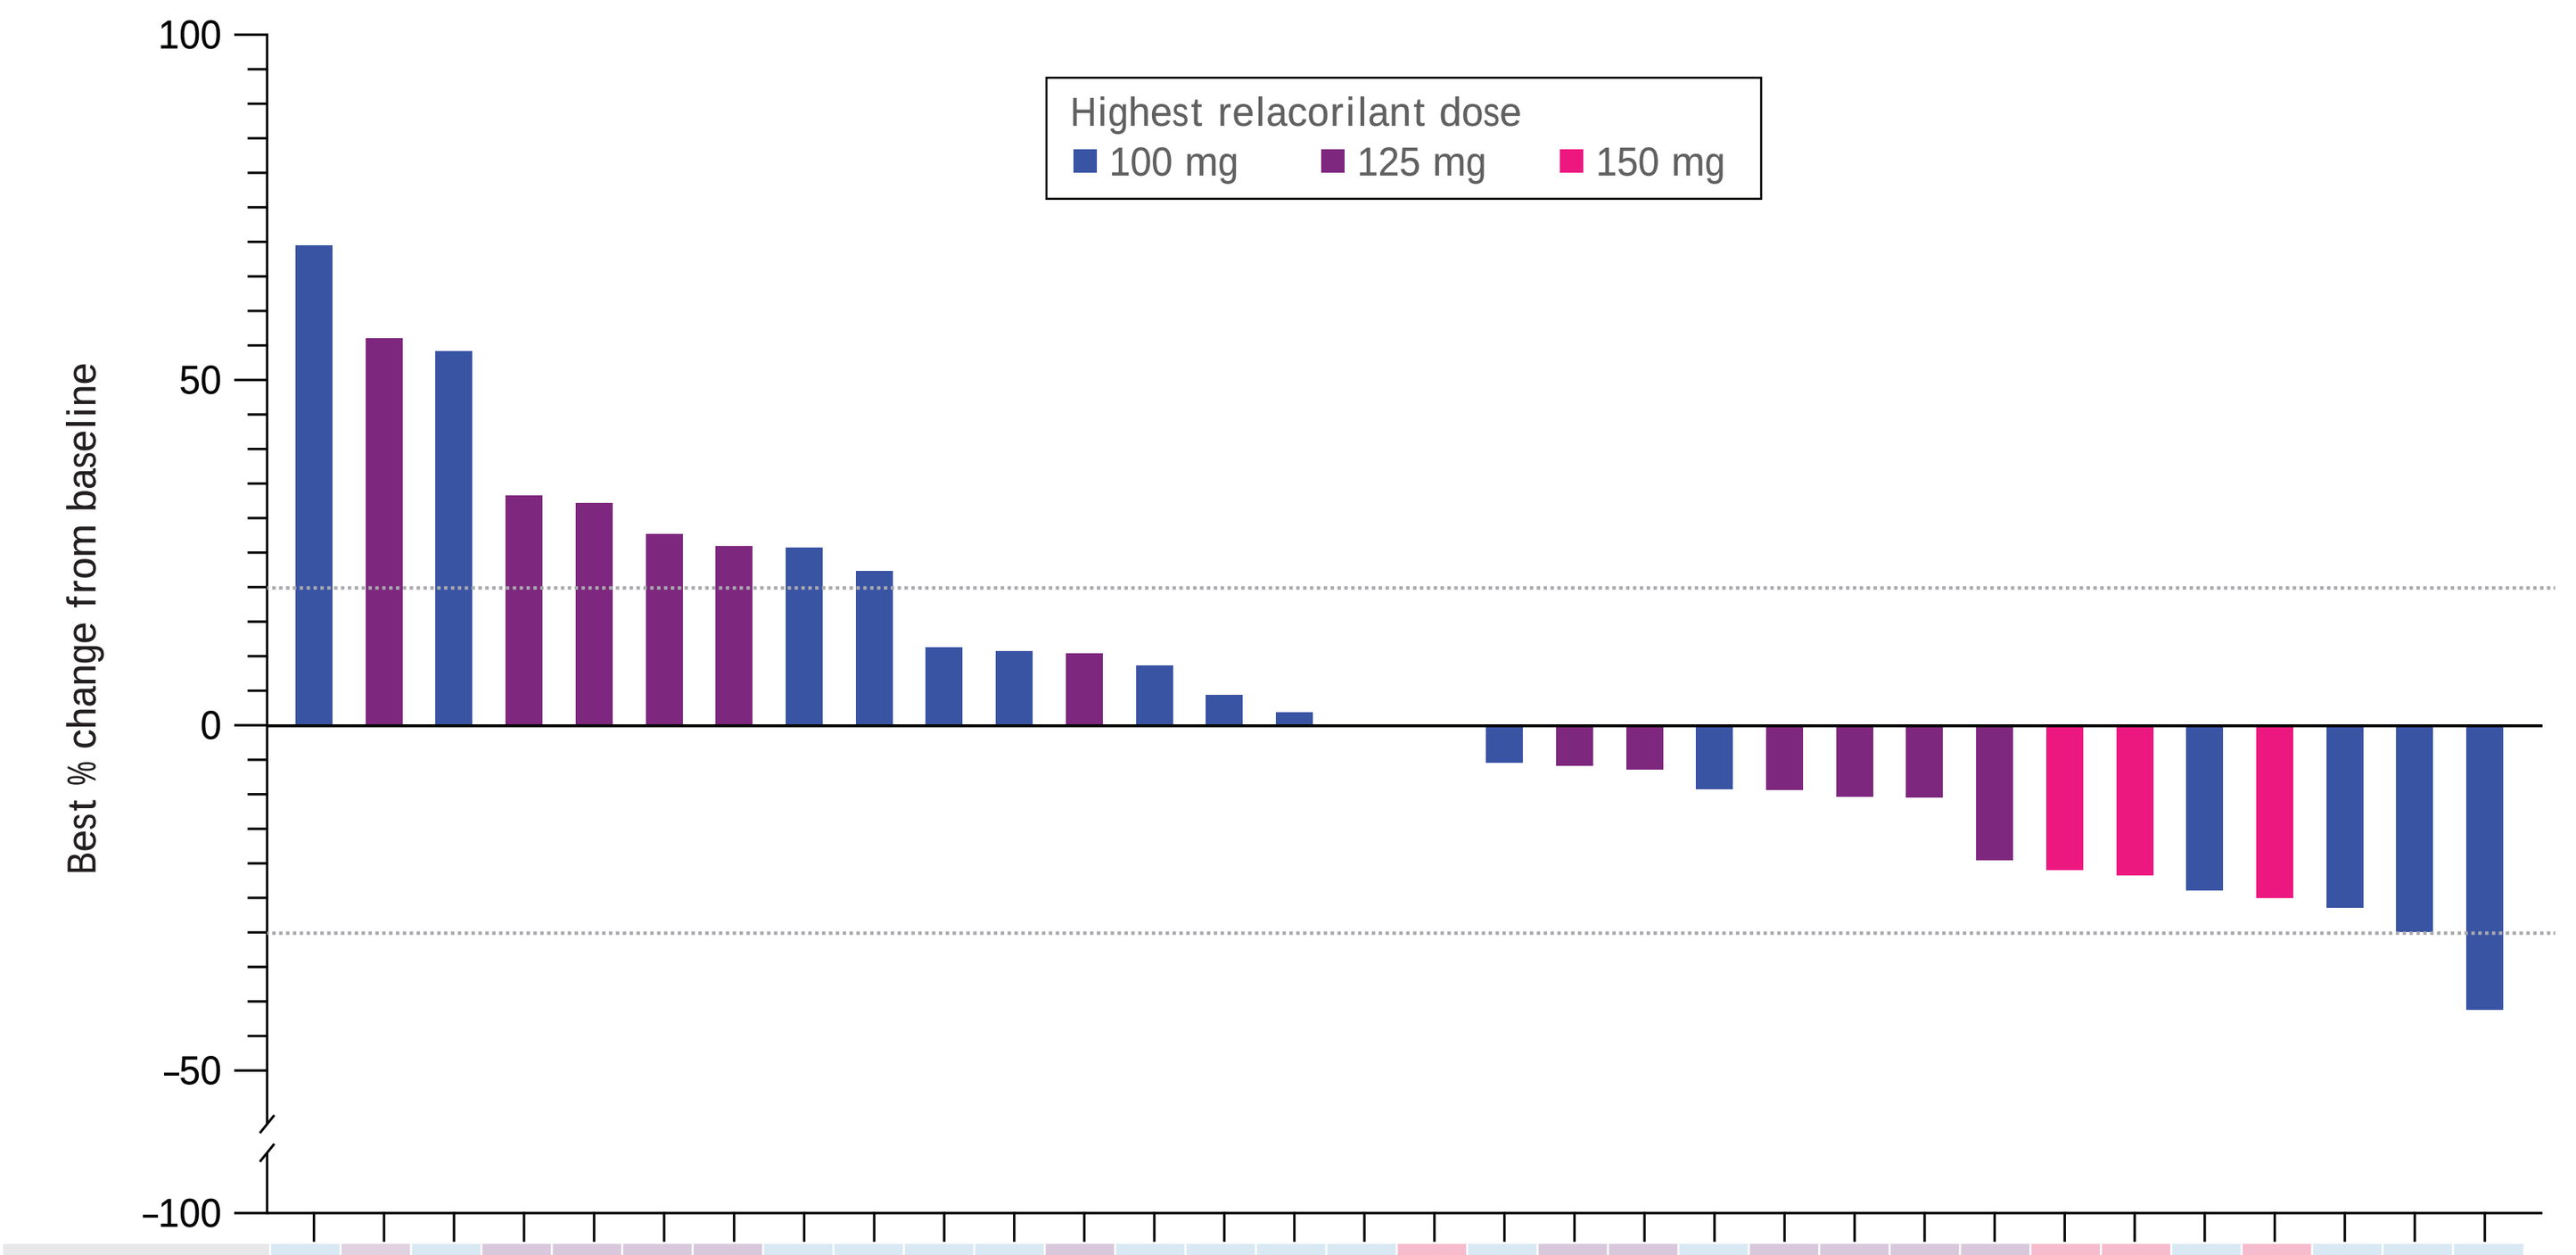

Highest relacorilant dose
100 mg
125 mg
150 mg
Best % change from baseline
100
50
0
–50
–100

Baseline KPS	90	80	90	80	70	90	80	80	70	90	70	90	80	70	80	90	90	70	90	80	90	70	90	80	80	90	80	90	80	70	80	80
No. of prior lines	2	2	4	2	2	2	2	2	3	3	4	3	3	2	4	3	3	2	3	3	4	4	2	3	4	2	2	3	3	2	3	3
Prior nab-paclitaxel	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	N	N	Y	Y	Y	Y	Y	Y	Y
Lung lesion	N	N	N	N	Y	Y	N	Y	N	Y	Y	Y	Y	Y	N	Y	Y	N	Y	Y	N	Y	Y	Y	Y	N	Y	Y	Y	Y	N	Y
Liver lesion	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	N	N	Y	Y	N	N	Y	Y	Y	Y	Y	Y	Y	N	Y	Y	N	Y	Y	Y	N
Baseline CA19-9	+	+	–	–	+	+	+	+	–	+	+	+	+	+	+	+	+	+	+	+	–	+	+	+	+	+	+	–	+	+	+	+

+, CA19-9 ≥40 U/mL; –, CA19-9 <40 U/mL.
KPS, Karnofsky performance status.

4 Safety Profile of Relacorilant + Nab-paclitaxel

Incidence of treatment-emergent AEs (≥20%)

Preferred term, n (%)	Relacorilant + nab-paclitaxel (N=43)
Fatigue	35 (81.4)
Nausea	21 (48.8)
Decreased appetite	17 (39.5)
Vomiting	15 (34.9)
Anemia	14 (32.6)
Hypoalbuminemia	12 (27.9)
Neutrophil count decreased	12 (27.9)
Abdominal pain	11 (25.6)
Back pain	11 (25.6)
Diarrhea	10 (23.3)
Hypokalemia	10 (23.3)
Hyponatremia	9 (20.9)
Peripheral edema	9 (20.9)
White blood cell count decreased	9 (20.9)

MedDRA version: 23.0.

- All patients (n=43) reported ≥1 treatment-emergent AE.
 - Grade ≥3 AEs were experienced by 37 patients (86.1%).
 - AE that led to a fatal outcome was reported for 3 patients (7.0%).
 - Of note, 19 patients (44.2%) experienced neutropenia or decreased neutrophil count.
- Of the 43 patients, 6 (14.0%) had ≥1 serious AE related to relacorilant.
 - Serious AEs included fever, fatigue, febrile neutropenia, pneumonitis, failure to thrive, diarrhea, vomiting, cytopenia, abdominal pain, and ascites.
 - Cases were confounded by nab-paclitaxel treatment and underlying disease and most did not lead to relacorilant dose modification.
- Overall, the safety profile of relacorilant + nab-paclitaxel observed in this study is consistent with that from previous relacorilant studies in oncology.[9,10]

5 Suppression of GR Target Genes Observed

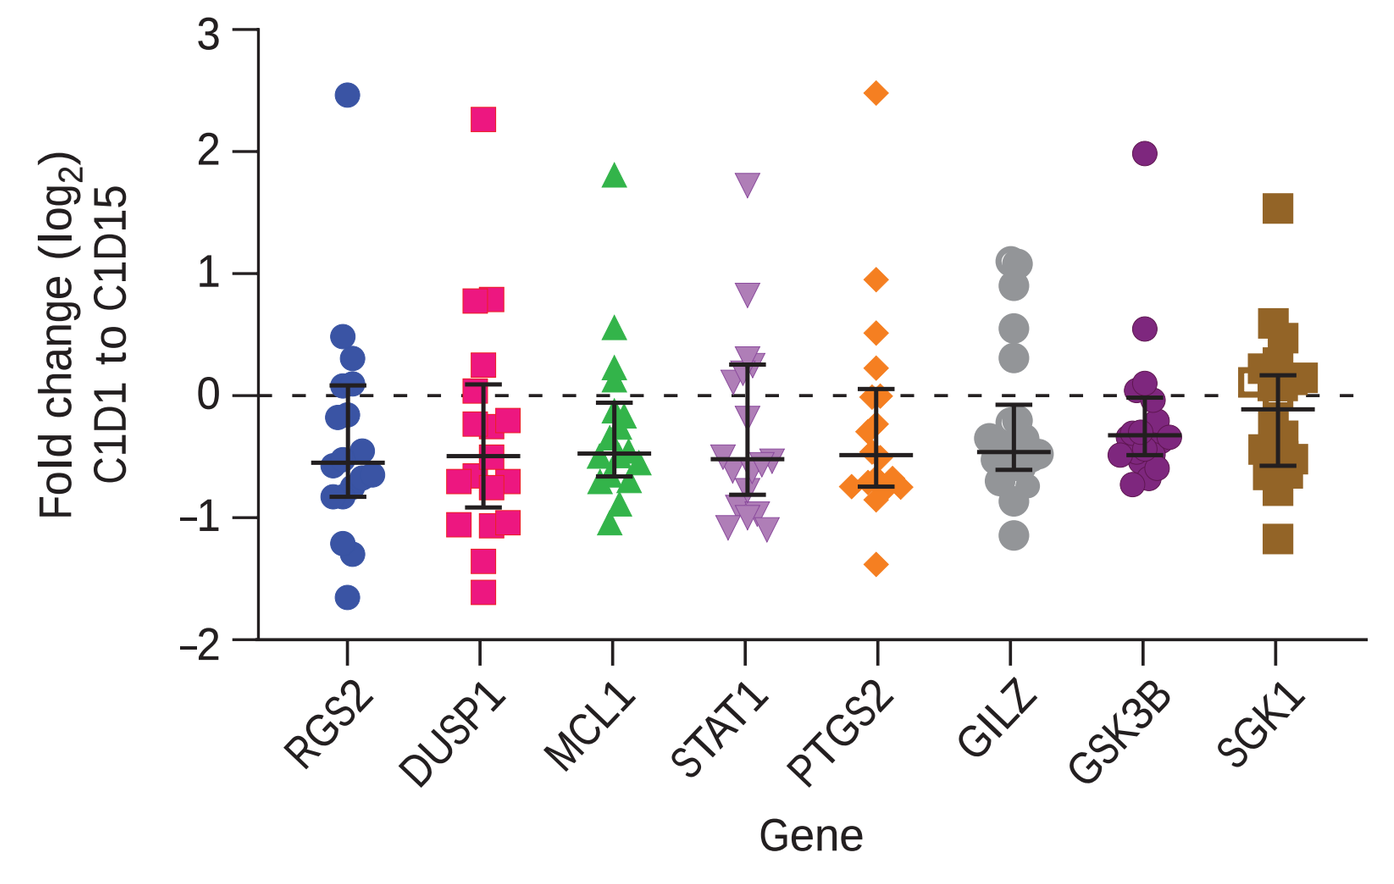

Fold change (log2) C1D1 to C1D15
3
2
1
0
–1
–2
RGS2
DUSP1
MCL1
STAT1
PTGS2
GILZ
GSK3B
SGK1
Gene

- Genes upregulated in response to a GR agonist (prednisone) in whole blood were identified in a separate study.
- These genes were suppressed by relacorilant + nab-paclitaxel.*

*Patients that received prohibited concomitant medications were not included in this analysis.
C, cycle; D, day.

6 Reduction in Tumor Size Observed by CT and FDG PET

Patient CT and FDG PET scans after treatment with relacorilant + nab-paclitaxel

Patient 1:

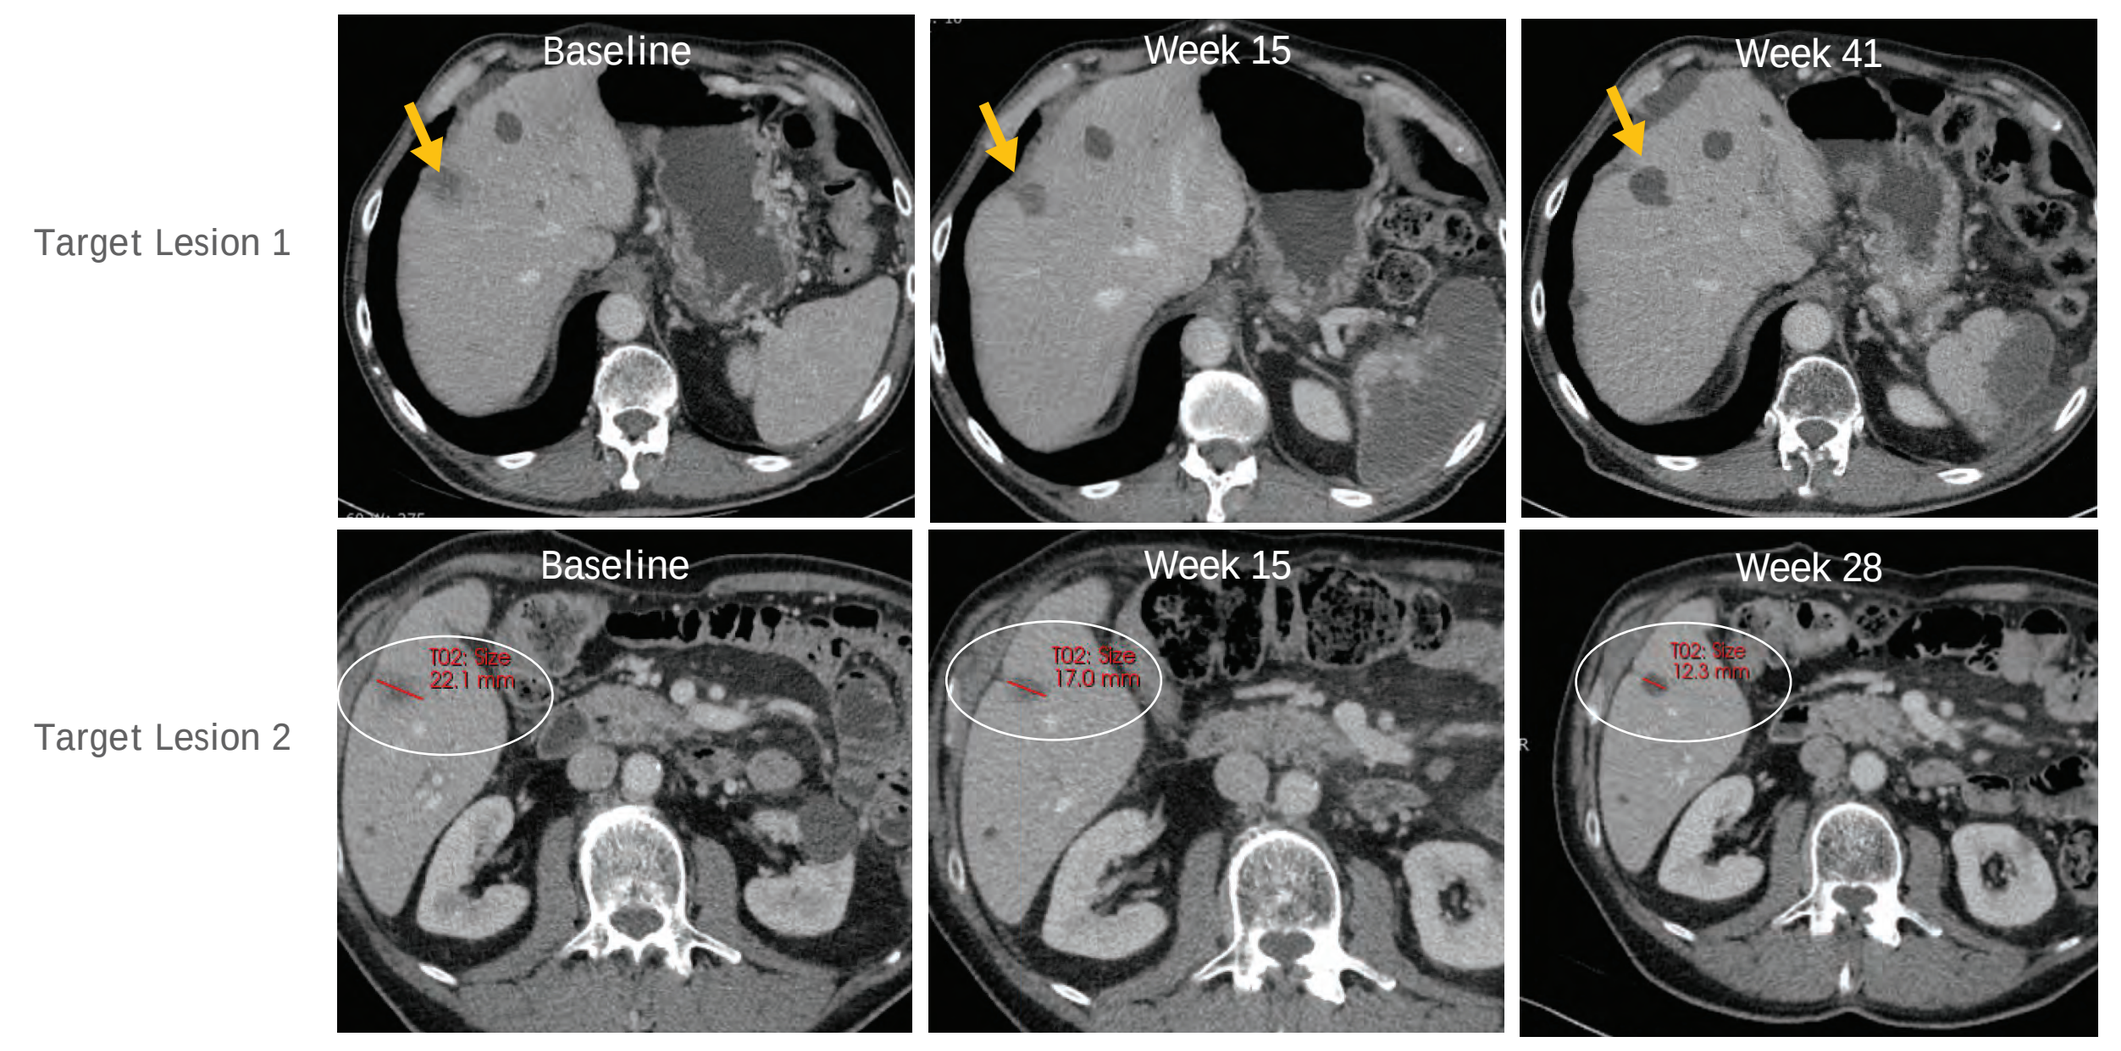

Target Lesion 1
Target Lesion 2
Baseline
Week 15
Week 41
Baseline
Week 15
Week 28

73-year-old male patient with 3 prior lines of therapy, including nab-paclitaxel. Patient was on treatment for 9 cycles. Multiple heterogeneously appearing hepatic metastases are present at baseline (yellow arrow and white circle). The metastatic lesions demonstrate tumor shrinkage and necrosis over time.

Patient 2:

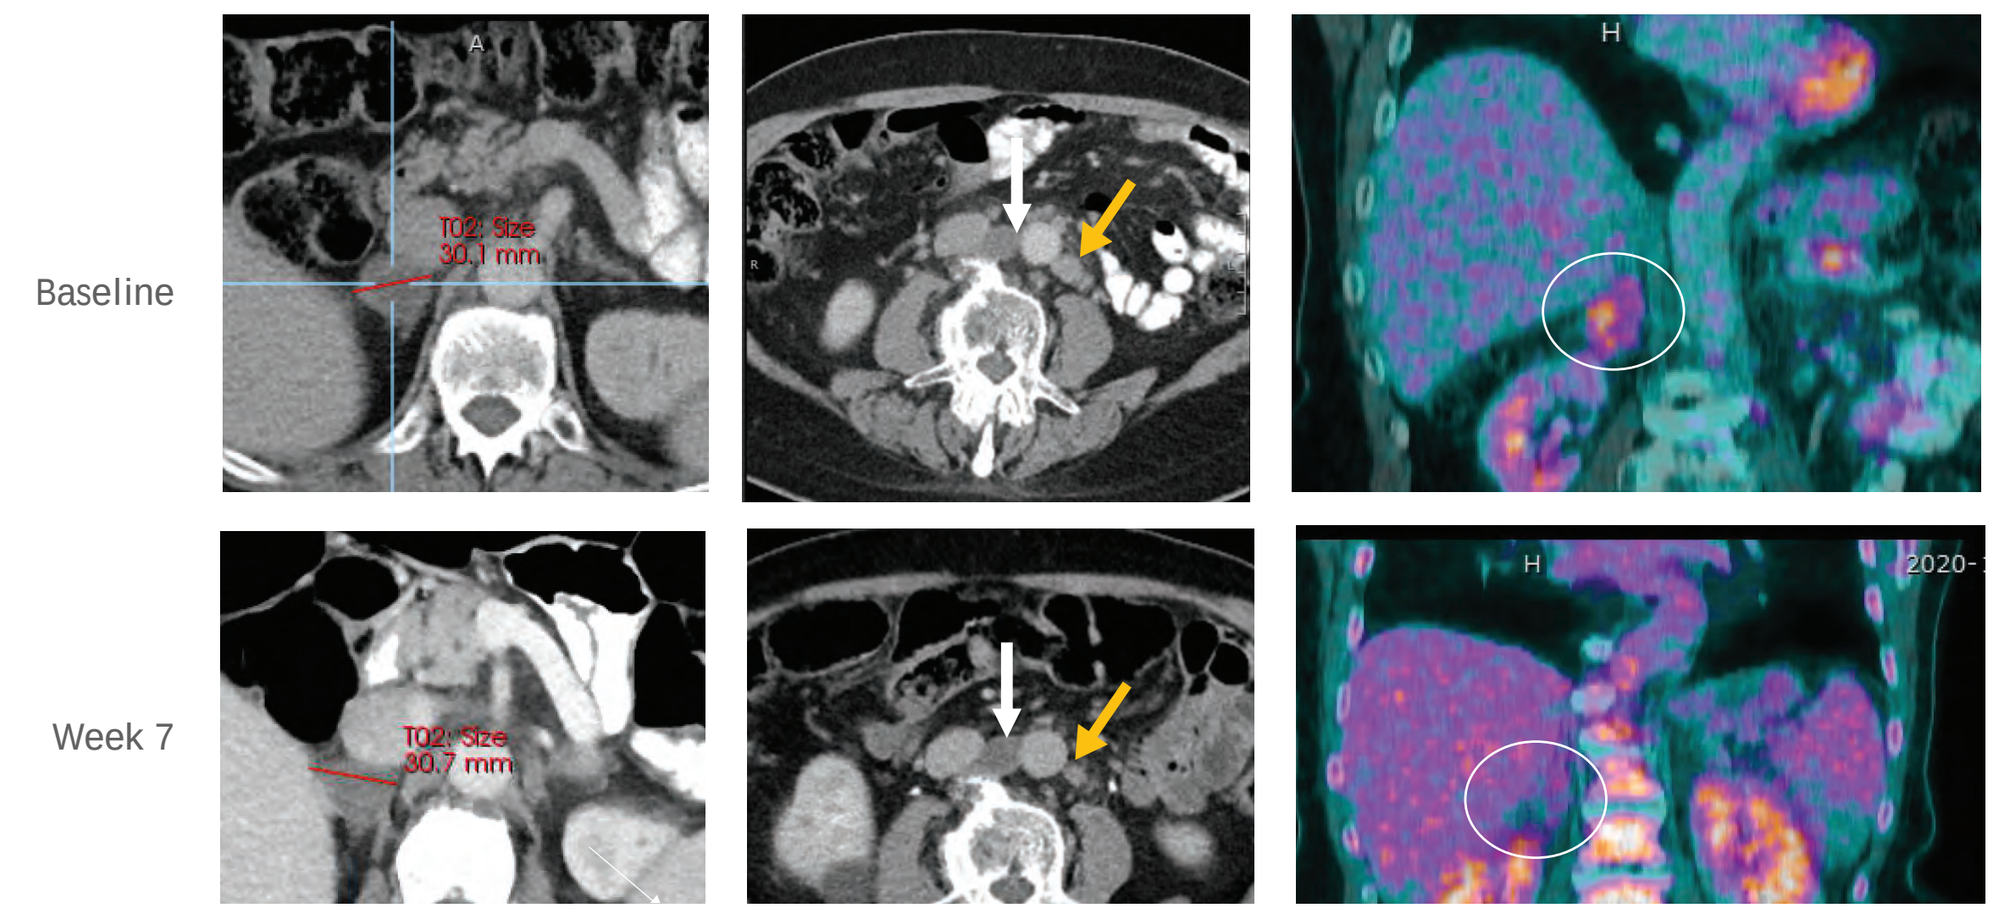

Baseline
Week 7

74-year-old male patient with 3 prior lines of therapy, including nab-paclitaxel. Patient was on treatment for 17 cycles. Metastatic disease, with FDG PET avid uptake involving the right adrenal gland and and several retroperitoneal lymphadenopathy, was noted at baseline. At Week 7, improvement in FDG PET with retroperitoneal lymph node stability and shrinkage was observed. (Yellow arrow = left periaortic lymph node, white arrow = aortocaval lymph node; white circle = right adrenal gland.)

CT, computed tomography; FDG PET, fluorodeoxyglucose positron emission tomography.

References

(1) Bray F, et al. *CA Cancer J Clin*. 2018;68(6):394-424.
(2) American Cancer Society. 2019. Cancer statistics center. https://cancerstatisticscenter.cancer.org/#!. Updated 2022. Accessed April 19, 2022.
(3) Pishvaian MJ, et al. *Lancet Oncol.* 2020;21(4):508-518.
(4) Du J, et al. *Bioscience Rep*. 2020;40(7):BSR20200401.
(5) Melhem A, et al. *Clin Cancer Res.* 2009;15(9):3196-3204.
(6) Zhang C, et al. *Int J Oncol*. 2006;29(5):1295-1301.
(7) Greenstein AE, Hunt HJ. *Oncotarget*. 2021;12(13):1243-1255.
(8) Custodio JM, et al. *J Clin Pharmacol*. 2021;61(2):244-253.
(9) Munster P, et al. Presented at: American Society of Clinical Oncology Annual Congress; May 31-June 4, 2019; Chicago, IL, USA. Poster 4130.
(10) Colombo N, et al. Presented at: European Society for Medical Oncology Annual Meeting; September 16-21, 2021; Paris, France; Abstract 721O.

Acknowledgments

This study was funded by Corcept Therapeutics. Medical writing was provided by Matt McErlean, PhD, and Claire E. Brady, PharmD (Cello Health Communications) and funded by Corcept Therapeutics. The authors reviewed and approved this poster.

Disclosures

Erkut H. Borazanci: Data safety monitoring board or advisory board for Corcept. **Nathan Bahary:** Consulting for Bristol Meyers Squibb, Pfizer, AstraZeneca. **Vincent Chung** and **Paul E. Oberstein:** No conflicts to disclose. **Roland T. Skeel:** Faculty of University of Toledo. **E. Gabriela Chiorean:** Grants or contracts for Corcept. **Andrew Greenstein:** Employee of Corcept; Stock options in Corcept; Patents for Corcept. **Hristina Pashova:** Employee of Corcept; Stock or stock options in Corcept. **Iulia Cristina Tudor:** Employee of Corcept; Stock options in Corcept. **Grace Mann:** Employee of Corcept; Stock options in Corcept.